The crystal structure of the RuvBL1/RuvBL2 complex: how to make the most of low resolution data

Pedro M. Matias[a,b], Tiago M. Bandeiras[b], Sabine Gorynia[c] , Filipa G. Pinho[b], Colin E. McVey[a] and Maria Arménia Carrondo[a]

[a]Instituto de Tecnologia Química e Biológica, Av. República, 2780 Oeiras, Portugal
[b]Instituto de Biologia Experimental e Tecnológica, Av. República, 2780 Oeiras, Portugal
[c]Bayer Schering Pharma, GDD-LGO-LDB-Protein Supply, Muellerstrasse 170-178, 13342 Berlin, Germany

Bayer HealthCare
Bayer Schering Pharma

1. INTRODUCTION

RuvBL1 (RuvB-like) and its homolog RuvBL2 are evolutionarily highly conserved eukaryotic proteins belonging to the AAA+ family of ATPases (ATPases associated with diverse cellular activities) (Neuwald et al., 1999). They were found to be present in diverse chromatin remodeling complexes, which regulate chromatin structure and access of proteins to DNA. RuvBL1 and RuvBL2 regulate transcription not only via association with chromatin remodeling complexes, but also through interactions with diverse transcription factors and the RNA polymerase II holoenzyme complex.

RuvBL1 and RuvBL2, consisting of 456 and 463 amino acids respectively, exhibit 43 % identity and 65 % similarity. The crystal structure of RuvBL1 was determined in 2006 in our laboratory (Matias et al., 2006).

2. EXPRESSION AND PURIFICATION

For crystallization purposes the domain II of both RuvBL1 and RuvBL2 was truncated (RuvBL1ΔDII and RuvBL2ΔDII). 6xHis-tagged RuvBL1 and FLAG-tagged RuvBL2 were co-expressed in *E.coli* and purified in three steps using two affinity purifications and a gel filtration.

3. CRYSTALLIZATION

Initial crystals were obtained by hanging-drop vapor-diffusion, with a drop composition of 2 µl protein solution (20 mg/ml complex in 20 mM Tris-HCl pH 8.0, 200 mM NaCl, 5 % glycerol, 4 mM $MgCl_2$, 2 mM β-mercaptoethanol) and 2 µl reservoir solution, equilibrated against 500 µl of precipitant solution in the well. The best diffracting crystals were obtained with a reservoir solution of 0.8 M LiCl, 10 % PEG 6000 and 0.1 M Tris pH 7.5. One crystal obtained under these conditions diffracted to 4 Å resolution and was used to measure diffraction data leading to a preliminary structure determination. The crystal was a fragment of a thin (*ca.* 20 µm) hexagonal-shaped plate.

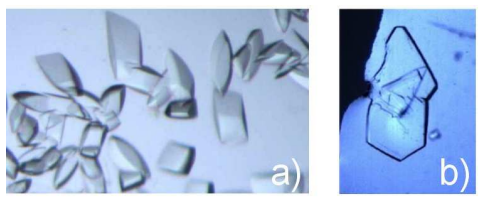
a)
b)

Figure 1. RuvBL1ΔDII/RuvBL2ΔDII complex crystals and diffraction pattern. **a)** RuvBL1ΔDII/RuvBL2ΔDII crystals; **b)** optimized hexagonal-shaped plates used for preliminary structure determination.

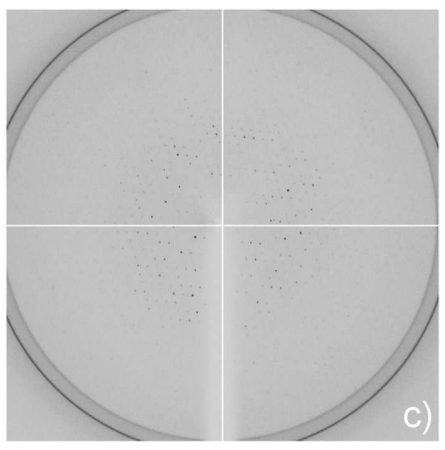
c)

c) Diffraction image to 4 Å resolution. The ice rings surrounding the diffraction pattern may be due to accidental thawing and freezing of the crystal in the loop and may prevent seeing spots at a slightly higher resolution of about 3.5 Å.

4. DATA COLLECTION

Diffraction data were collected at the European Synchrotron Radiation Facility (ESRF) in Grenoble and were integrated to 4 Å resolution with XDS (Kabsch, 1993) and further processed with SCALA and TRUNCATE in the CCP4 suite (Collaborative Computational Project Number 4, 1994).

The diffraction pattern could be indexed and integrated in two possible space groups:

- Orthorhombic $C222_1$ with cell parameters a=111.4, b=188.0, c=243.4 Å and 6 monomers in the asymmetric unit, R_{merge}=14%, $\langle I/\sigma(I)\rangle$=3.7.
- Monoclinic $P2_1$ with cell parameters a=109.2, b=243.4, c=109.3 Å, β=118.7° and 12 monomers in the asymmetric unit., R_{merge}=12%, $\langle I/\sigma(I)\rangle$=3.5.

5. STRUCTURE DETERMINATION

The 3D structure of the RuvBL1ΔDII/RuvBL2ΔDII complex was solved in both possible space groups by the Molecular Replacement method using the program PHASER (Storoni *et al.*, 2004). The search model was the homologous RuvBL1 monomer (Matias *et al.*, 2006), truncated to reflect the shortened domain II region.

Solution obtained: a **dodecamer** formed by two hexamers.

In $P2_1$ a full dodecamer constitutes the asymmetric unit; in $C222_1$ only one hexamer is contained in the asymmetric unit (**Figure 2**).

The high similarity between the 3D structures of the RuvBL1ΔDII and RuvBL2ΔDII combined with the low data resolution, made rather difficult the distinction between RuvBL1 and RuvBL2 monomers, as well as between space groups $C222_1$ and $P2_1$.

§ Present Address: Oncotest GmbH - Institute for Experimental Oncology, Am Flughafen 12-14, 79108 Freiburg Germany

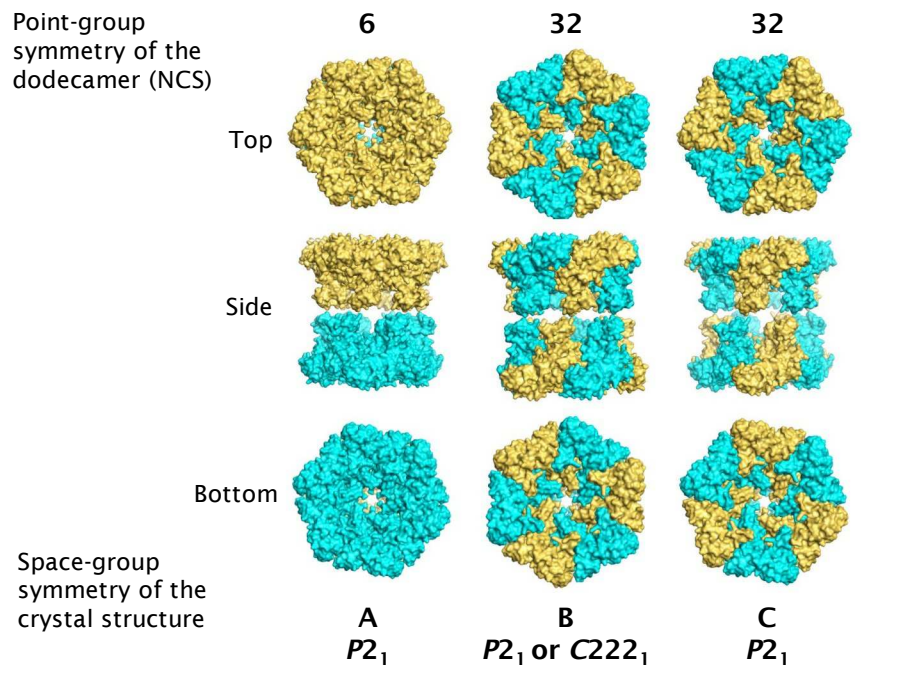
Point-group symmetry of the dodecamer (NCS)
6
32
32
Top
Side
Bottom
Space-group symmetry of the crystal structure
A
P2₁
B
P2₁ or C222₁
C
P2₁

Figure 2. The three possible structures for the RuvBL1ΔDII/RuvBL2ΔDII complex in space groups $P2_1$ and $C222_1$: One dodecamer formed by two homohexamers (**A**) or two heterohexamers (**B** and **C**).

Self-rotation calculations with CCP4 MOLREP (**Figure 3**) appear to support a double heterohexamer in $C222_1$: the peaks in the κ=60, 120 and 180º sections are much stronger than in $P2_1$, and the peaks in the κ=120º section are stronger than those in the κ=60º.

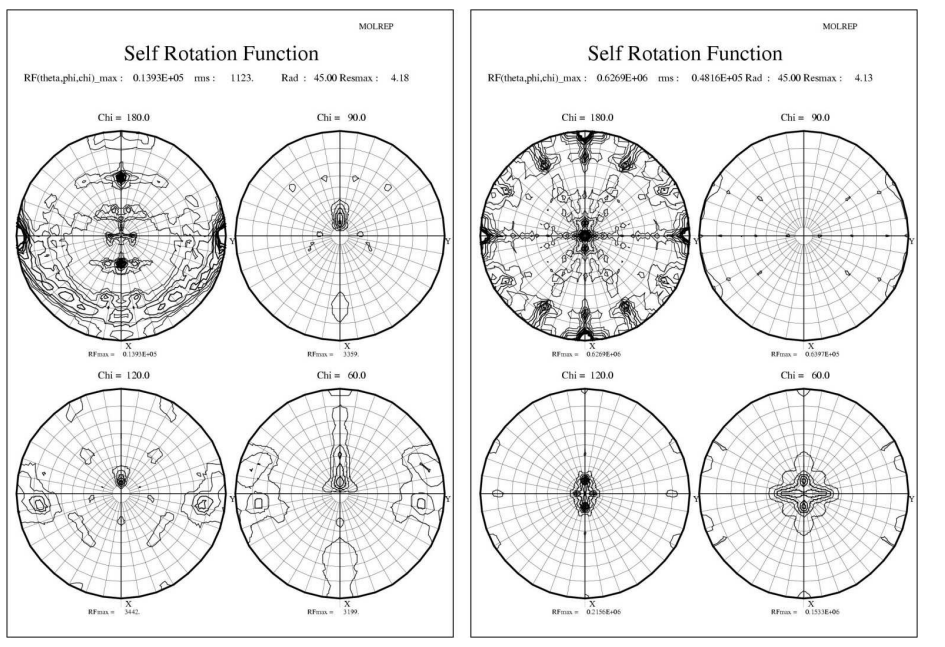
Self Rotation Function
MOLREP
Chi = 180.0
Chi = 90.0
Chi = 120.0
Chi = 60.0

Figure 3. Self-rotation function calculations with MOLREP in space groups $P2_1$ (left) and $C222_1$ (right). The contour levels are drawn at unit intervals between 1 and 6 map r.m.s. units. In $P2_1$, the crystallographic 2-fold along unit cell axis *b* can be seen as the strong peaks at the sides of the κ=180° section in the left panel. This axis corresponds to the crystallographic *c*-axis in $C222_1$, which is at the centre of the κ=180° section in the right panel. The strong peaks along the vertical axis on the $P2_1$ κ=180° section represent non-crystallographic 2-fold axes in $P2_1$ which become crystallographic in $C222_1$, along unit cell directions *a* and *b*.

Density calculations with DM (Cowtan, 1994) relying on NCS averaging (**Table 1**) also indicated a double heterohexamer in $C222_1$ to be the correct solution. However, this result contradicts all previous structural work based on electron microscopy of human RuvBL1/RuvBL2 complex and its Yeast homologue Rvb1/Rvb2 (**Figure 4**).

Table 1. Density Modification Results

Dodecamer in Figure 2	A	B	**B**	C
Space Group	$P2_1$	$P2_1$	$\mathbf{C222_1}$	$P2_1$
Final FOM from DM	0.754	0.783	**0.849**	0.757
Initial (final) NCS correlation between related density regions				
Domain 1	0.635 (0.835)	0.688 (0.862)	0.678 (**0.895**)	0.633 (0.834)
Domain 2	0.633 (0.832)	0.678 (0.861)	0.669 (**0.889**)	0.628 (0.830)

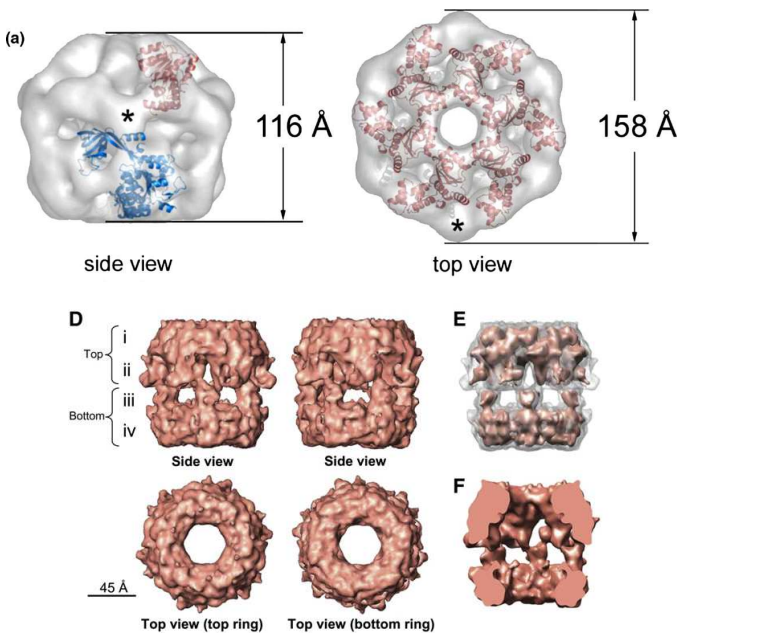
(a)
116 Å
158 Å
side view
top view
D
Top
Bottom
i
ii
iii
iv
Side view
Side view
E
F
45 Å
Top view (top ring)
Top view (bottom ring)

Figure 4. Top: Human complex, 20 Å resolution, asymmetric dodecamer, possibly two homohexamers facing each other (Puri *et al.,* 2007). **Bottom:** Yeast complex, 13 Å resolution, asymmetric dodecamer, possibly two homohexamers facing each other (Torreira et al., 2008). **Not shown:** yeast complex, heterohexamers, probably made up of alternating RuvBL1 and RuvBL2 monomers (Gribun et al., 2008).

6. RESOLVING THE AMBIGUITY

RuvBL1ΔDII and RuvBL2ΔDII each contain **11 methionine** residues, and with one exception they occupy different locations in the sequence (**Figure 5**).

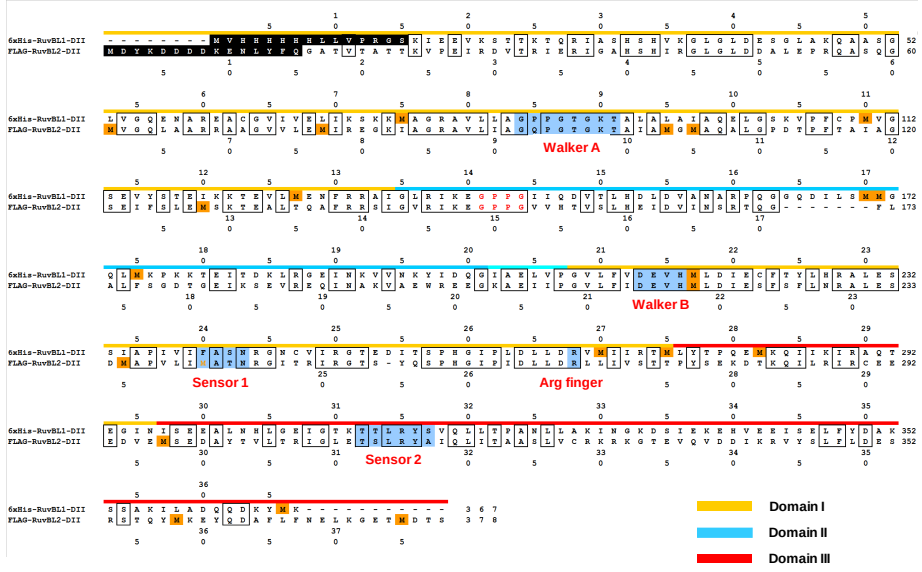
6xHis-RuvBL1-DII
FLAG-RuvBL2-DII
Walker A
Walker B
Sensor 1
Arg finger
Sensor 2
Domain I
Domain II
Domain III

Figure 5. Sequence alignment of RuvBL1ΔDII and RuvBL2ΔDII showing the domain and AAA+ regions, and the location of the methionine residues.

In order to elucidate the dodecamer composition by X-ray crystallography, the expression, purification and crystallization of the Se-Met derivative of the complex was undertaken (**Figure 6**).

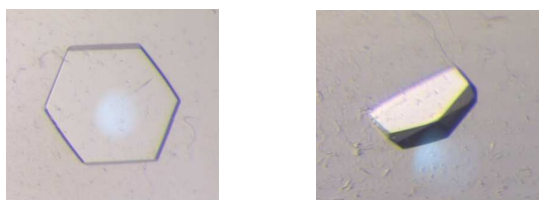

Figure 6. crystals of the Se-Met RuvBL1ΔDII/RuvBL2ΔDII complex were obtained at 4°C by sitting-drop vapor-diffusion, using a protein concentration of 12 mg/mL and 20 mM Tris-HCl pH 8.0, 200 mM NaCl, 10 % glycerol, 4 mM MgCl2, 4 mM ADP, 0.5 mM TCEP as the precipitating solution.

The structure was determined from a 3-wavelength MAD data set collected at the European Synchrotron Radiation Facility (ESRF) in Grenoble to a maximum resolution of 3 Å. The space group was unambiguously determined as $C222_1$ and the structure was refined with BUSTER (Bricogne et al., 2010) to R and R_{free} values of 0.178 and 0.205 respectively. The new results **confirmed** those previously obtained at 4 Å: The complex crystallizes as a dodecamer formed by two heterohexamers with **alternating** RuvBL1ΔDII and RuvBL2ΔDII monomers. One heterohexamer is present in the asymmetric unit of space group $C222_1$, and the second is generated by a crystallographic 2-fold rotation axis (**Figure 7**).

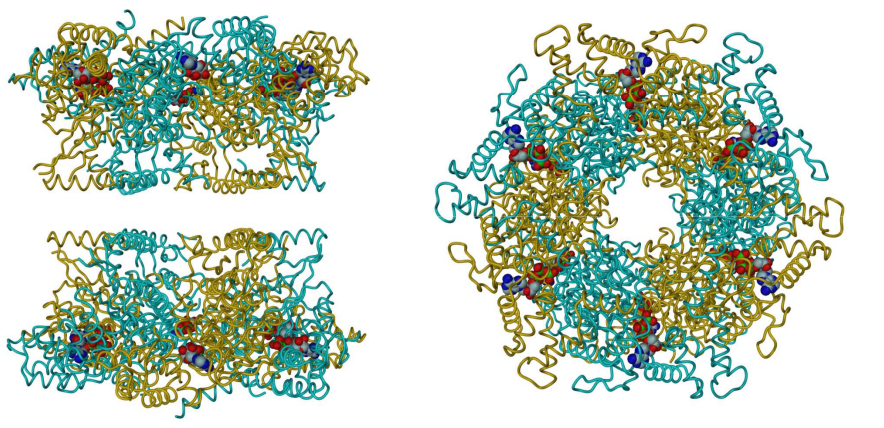

Figure 7. Side (left) and top (right) views of the RuvBL1ΔDII/RuvBL2ΔDII complex. RuvBL1ΔDIIand RuvBL2ΔDII monomers are drawn as tube C^{α} diagrams and are colored gold and cyan, respectively. ATP molecules are drawn as space-filling with atom colors light blue for carbon, red for oxygen, blue for nitrogen and green for phosphorus.

This poster is based on the publications: Gorynia, S., Matias, P.M. Bandeiras, T.M., Donner, P & Carrondo, M.A. (2008) *Acta Crystallogr.* F64:840-846 and Gorynia, S., Bandeiras, T.M., Pinho, F.G., McVey, C.E., Vonrhein, C., Round, A., Svergun, D. I., Donner, P., Matias, P. M. & Carrondo, M.A. (2011) *J. Struct. Biol.*, 176:279-291.

Acknowledgements: This work was supported by Bayer Schering Pharma (Berlin), Fundação para a Ciência e Tecnologia (Portugal) grant PEst-OE/EQB/LA0004/2011 and European Commission funding through the SPINE2COMPLEXES project LSHGCT2006031220. We also thank the ESRF for support with the data collections.

REFERENCES

- Bricogne, G., Blanc, E., Brandl, M., Flensburg, C., Keller, P., et al. (2010) BUSTER version 2.9. Global Phasing Ltd., Cambridge, UK.
- Collaborative Computational Project Number 4 (1994) *Acta Crystallogr.* D50:760-763.
- Cowtan, K. (1994) in: Joint CCP4 and ESF-EACMB Newsletter on Protein Crystallography Vol. 31, Daresbury Laboratory, Warrington, U.K., pp. 34-38
- Gribun, A., Cheung, K. L., Huen, J., Ortega, J. & Houry, W. A. (2008). *J. Mol. Biol.* 376:1320-1333.
- Kabsch, W. (1993) *J. Appl. Crystallogr.* 26:795-800.
- Matias, P. M., Gorynia, S., Donner, P. & Carrondo, M. A. (2006) *J. Biol. Chem.* 281:38918-38929.
- Neuwald, A. F., Aravind, L., Spouge, J. L. & Koonin, E. V. (1999) *Genome Res. 9*:27-43.
- Puri, T., Wendler, P., Sigala, B., Saibil, H. & Tsaneva, I. R. (2007) *J. Mol. Biol.* 366:179-192.
- Storoni, L.C., McCoy, A.J. & Read, R.J. (2004) *Acta Crystallogr.* D60:432-438.
- Torreira, E., Jha, S., Lopez-Blanco, J.R., Arias-Palomo, E., Chacon, P., et al., (2008) *Structure* 16:1511-1120.